Utility of speech-based digital biomarkers for evaluating disease progression in clinical trials of Alzheimer's disease

William Simpson, PhD [1,2], Liam D. Kaufman, MSc [2], Mike Detke, MD PhD [3], Casey Lynch, MSc [3], Adam Butler BA [2], Steve Dominy, MD [3]

(1) Department of Psychiatry and Behavioural Neuroscience, McMaster University, Hamilton, ON, Canada, (2) Winterlight Labs, Toronto, ON, Canada, (3) Cortexyme Inc, South San Francisco, CA, USA.

Background

Recent clinical trials in Alzheimer's disease have been overwhelmingly negative, spurring development of novel biomarkers which might capture changes in cognitive function with greater precision. Computational analysis of speech and language represent one such group of biomarkers (1,2). The objective of this study was to examine the utility of a speech-based digital biomarker for tracking disease progression and treatment response to investigational treatment COR388 in a group of patients with Alzheimer's disease.

Methods

- This study was a small, double-blind placebo-controlled Phase 1b trial (NCT03418688) of COR388 with a cohort of nine individuals with Alzheimer's disease.
- Participants were randomized to receive COR388 (6) or placebo (3) in a 2:1 fashion, BID for 28 days.
- A tablet-based speech and language assessment was administered at Screening, Days 1, 15 and 28. Participants were asked to complete 2 picture description tasks and verbal responses were analyzed
- Using natural language processing techniques, audio samples and transcripts were analyzed to generate 3 experimental composite metrics, chosen for their previous association to AD: syntactic complexity, information units and word finding difficulty (WFD).

Results

- Previous analysis of single outcomes showed significant improvement in the quality of picture descriptions for COR388 patients relative to placebo (increase in unique object content units, p=0.016 and prepositions, p=0.0011) (3).
- Positive trends, but no significant differences in MMSE scores were also observed.
- For experimental composite markers, mean baseline to endpoint comparisons showed trend level improvements in information units, syntactic complexity and word finding difficulty in those treated with COR388.
- No significant within-subject differences were observed for placebo on any of 3 metrics.

Figure 1: Baseline to endpoint change scores for MMSE, prepositions, information units, syntactic complexity and word finding difficulty

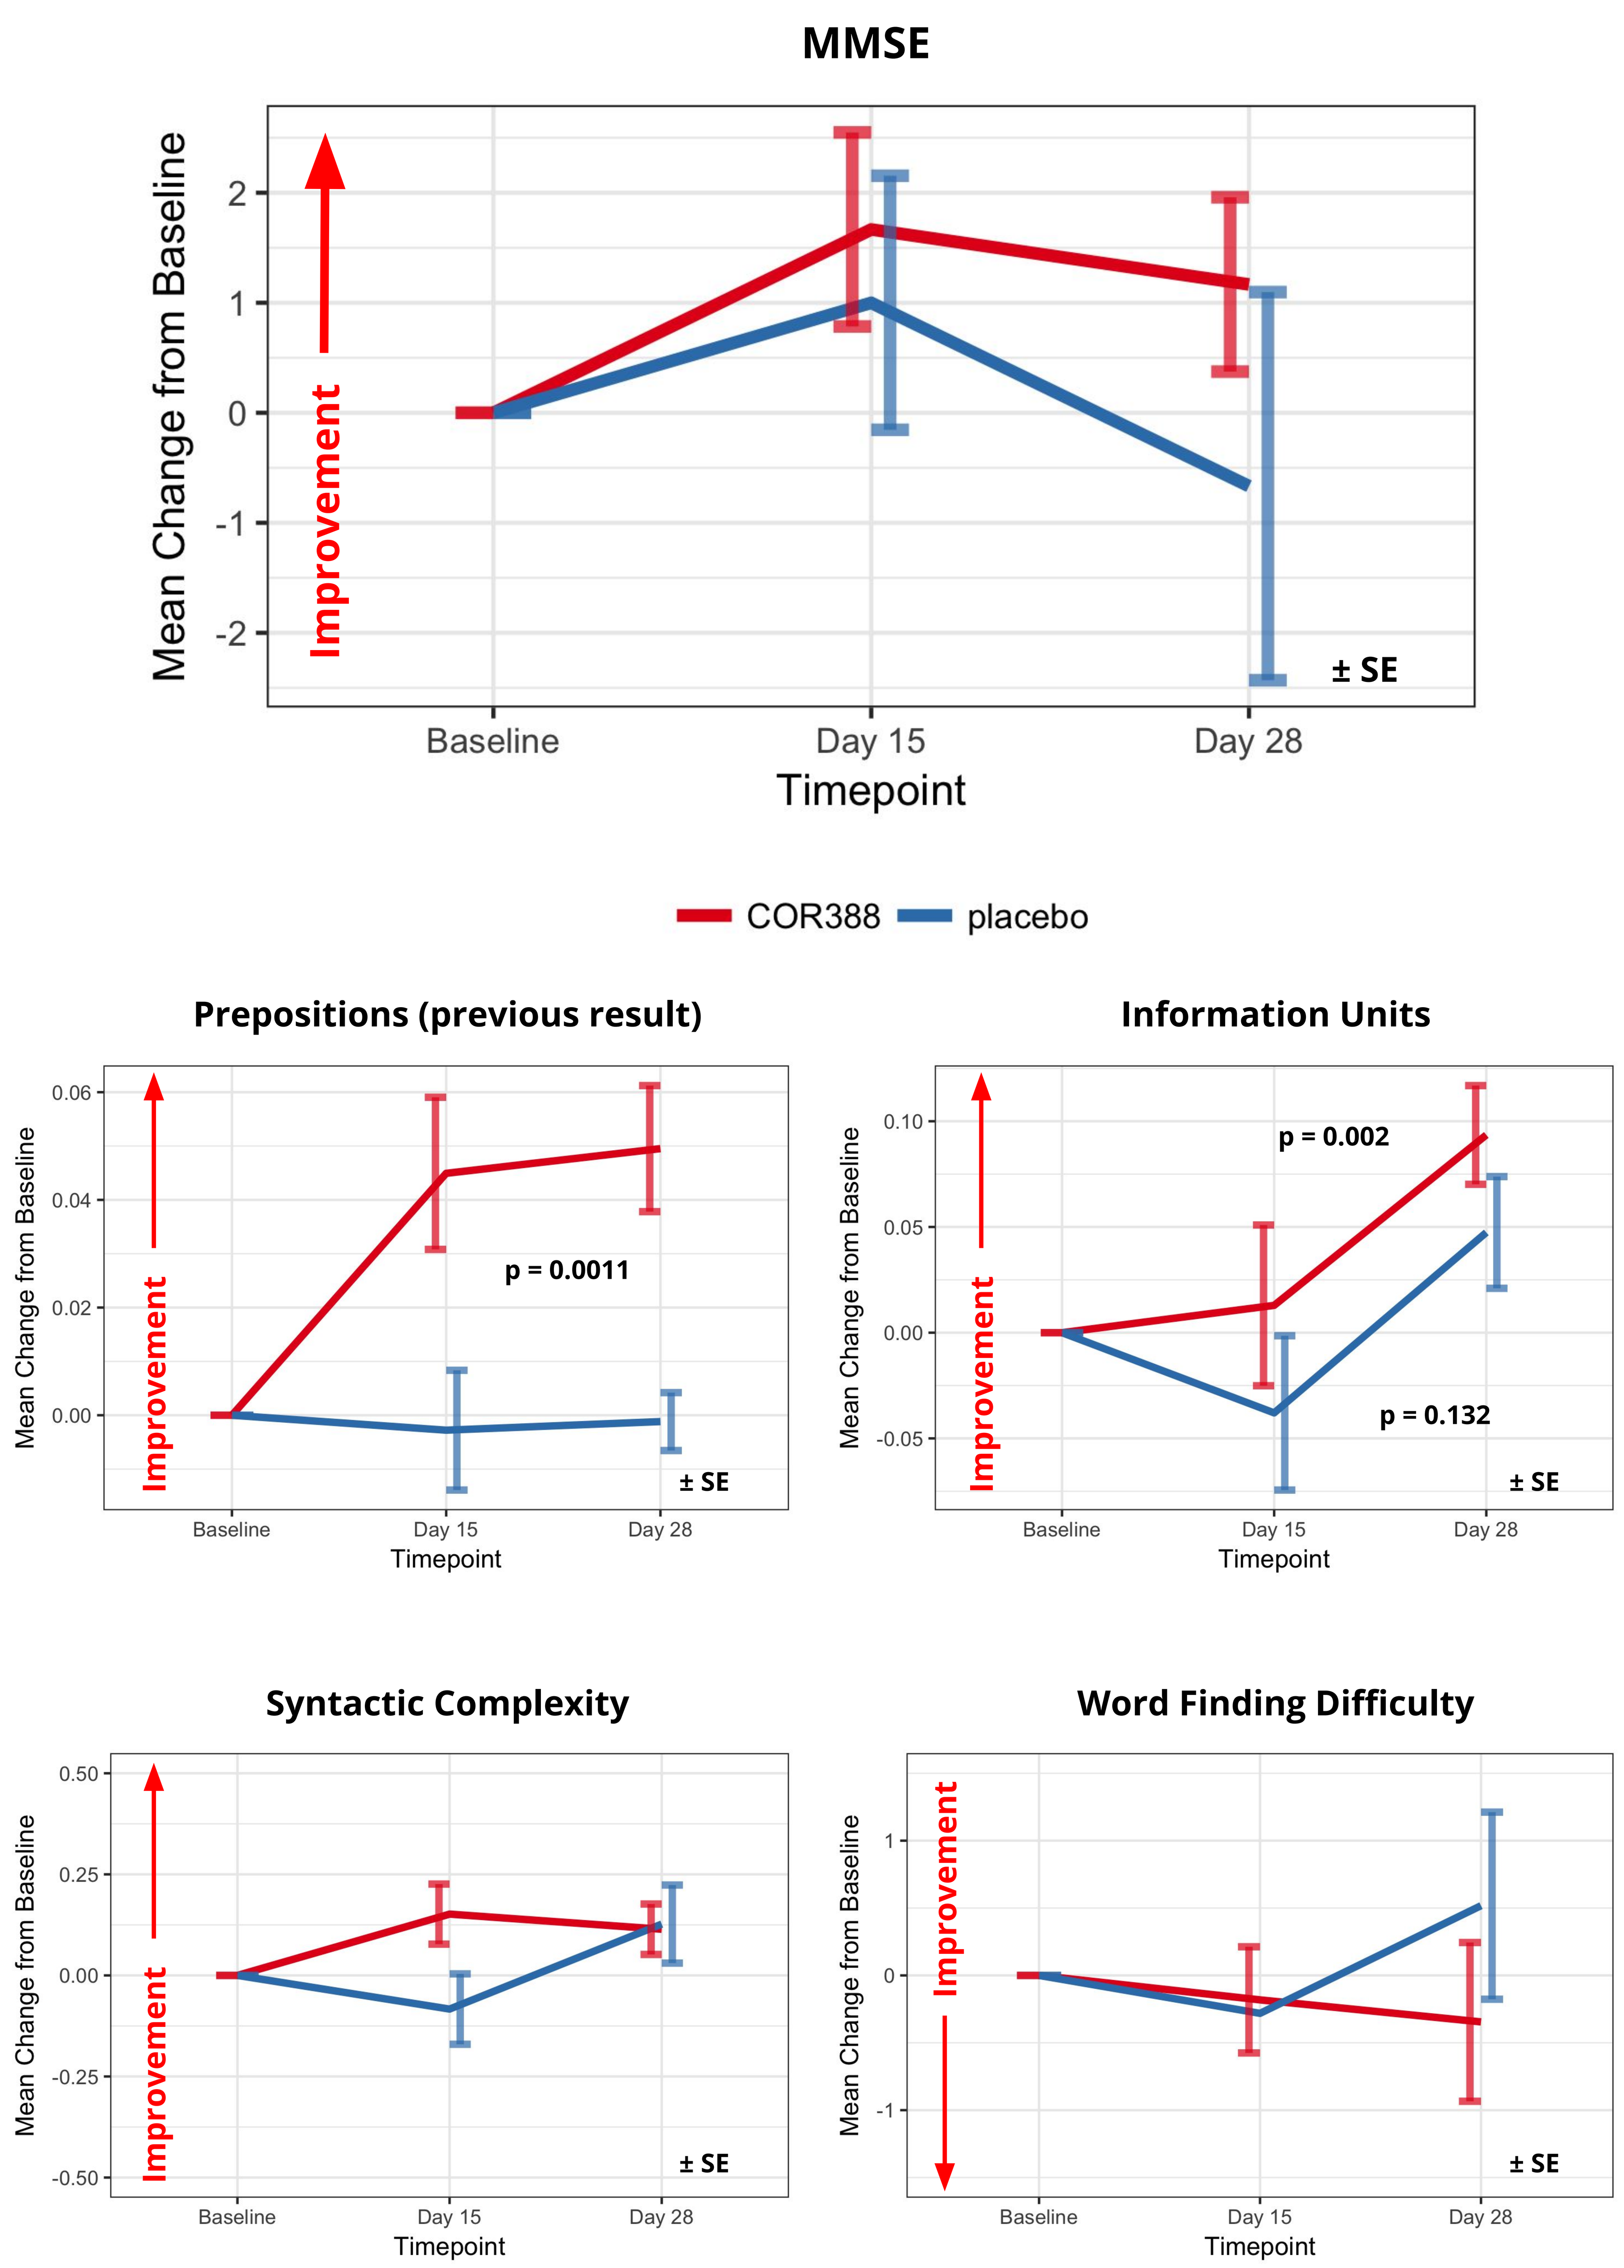
MMSE
Mean Change from Baseline
Improvement
± SE
Baseline
Day 15
Day 28
Timepoint
COR388
placebo
Prepositions (previous result)
p = 0.0011
Information Units
p = 0.002
p = 0.132
Syntactic Complexity
Word Finding Difficulty

Results Continued

- Baseline to endpoint, within-subject differences in information units were significant for COR388 participants, post Bonferroni correction (p=0.002).
- Between-group analysis of information units revealed a numerical but not statistical advantage for COR388 (~10pts v. ~5pts, p=0.21)
- For COR388 participants, there was also a positive trend indicative of improvement in Word Finding Difficulty.

Metric	Mean Change (Baseline-Day 28)		Within group p		Between group p
	COR388	PBO	COR388	PBO	
MMSE	1.67	-0.66	0.052	0.576	0.17
Information Units	0.093	0.047	**0.002**	0.132	0.21
Syntactic Complexity	0.114	0.126	0.094	0.245	0.91
Word Finding Difficulty	-0.345	0.517	0.570	0.245	0.36

(Bold) exceeds Bonferroni threshold p

Conclusions

- In this preliminary trial, patients treated with COR388 showed signs of significant improvement relative to placebo using a speech-based, digital biomarker.
- No significant changes in MMSE were observed suggesting that composite metrics derived from speech may represent sensitive tools for tracking changes in cognition in small trials.
- The study highlights the potential therapeutic benefit of COR388, though additional studies of sufficient power are needed.

References

1) Slegers, A., Filiou, R.-P., Montembeault, M. & Brambati, S. M. Connected Speech Features from Picture Description in Alzheimer's Disease: A Systematic Review. *J. Alzheimers. Dis.* 65, 519–542 (2018).

2) Fraser C, Meltzer A, and Rudzicz F. Linguistic Features Identify Alzheimer's Disease in Narrative Speech. Journal of Alzheimer's Disease: JAD 49 (2): 407–22, 2016.

3) Kaba S, Lynch C, Raha D, et al. A randomized, placebo controlled repeat dose Phase 1 study of COR388 in older healthy volunteers and patients with Alzheimer's disease. Poster: Presented at Clinical Trials for Alzheimer's Disease (CTAD) 2018. Barcelona, Spain, Oct 24-27, 2018.

Presented at Alzheimer's Association International Conference (AAIC) 2019, Los Angeles, CA, USA. July 14-18, 2019